PRESENTER ON BEHALF OF THE AUTHORS:
Mario Maldonado
(maldonado.m@recordati.com)

SESSION NO. **792**

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of this poster.

Alternatively, please visit the link:
https://qr.amiculum.biz/m0pyuy/

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from AACE and the authors of this poster.

Assessing Long-Term Safety and Efficacy of Osilodrostat in Prior- and New-Use Patients With Endogenous Cushing's Syndrome Enrolled in the Non-Interventional, Multinational LINC 6 Study: 2-Year Real-World Interim Analysis

Eliza B Geer,[1] Nicolas Scheyer,[2] Frédéric Castinetti,[3] Stephanie Espiard,[4] Celine Mouly,[5] Beverly MK Biller,[6] Maria Fleseriu,[7] Antoine Tabarin,[8] Martin Reincke,[9] Rosario Pivonello,[10] Richard Feelders,[11] Jeannie Le Mouhaër,[12] Peppi Raunioniemi,[13] Arnd Mueller,[13] Irina Bancos[14]

[1]Multidisciplinary Pituitary and Skull Base Tumor Center, Departments of Medicine and Neurosurgery, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Département d'Endocrinologie, Diabétologie et Nutrition, CHRU de Nancy, Hôpital Brabois et Université de Lorraine, Vandœuvre-lès-Nancy, France; [3]Aix-Marseille University, INSERM, MMG, Department of Endocrinology, La Conception Hospital, Assistance Publique Hôpitaux de Marseille, Marseille, France; [4]CHU de Lille, Lille, France; [5]Department of Endocrinology, CHU de Toulouse, Toulouse, France; [6]Neuroendocrine and Pituitary Tumor Clinical Center, Massachusetts General Hospital, Boston, MA, USA; [7]Pituitary Center, Departments of Medicine and Neurological Surgery, Oregon Health & Science University, Portland, OR, USA; [8]CHU de Bordeaux and Centre de Référence des Maladies Rares de la Surrénale, Bordeaux, France; [9]Ludwig-Maximilians-Universität München, Munich, Germany; [10]Dipartimento di Medicina Clinica e Chirurgia, Sezione di Endocrinologia, Università Federico II di Napoli, Naples, Italy; [11]Department of Internal Medicine, Endocrine Section, Erasmus Medical Center, Rotterdam, Netherlands; [12]Recordati Rare Diseases SARL Paris, France; [13]Recordati AG, Basel, Switzerland; [14]Division of Endocrinology, Metabolism and Nutrition, Mayo Clinic, Rochester, MN, USA

INTRODUCTION

- Endogenous Cushing's syndrome often requires lifelong medical treatment; further long-term data are therefore needed for all etiologies, including non-pituitary forms, of Cushing's syndrome
- The potent 11β-hydroxylase inhibitor osilodrostat provides rapid and sustained cortisol normalization and improvements in clinical signs and symptoms and quality-of-life outcomes in patients with Cushing's syndrome
 - Long-term safety and efficacy of osilodrostat were shown in the LINC clinical trial program in patients with Cushing's disease (Phase II: LINC 2, NCT01331239; Phase III: LINC 3, NCT02180217 and LINC 4, NCT02697734)[1–5]
 - Osilodrostat efficacy and safety were also evaluated in a Phase II study in Japanese patients with non-pituitary forms of Cushing's syndrome[6]
- The LINC 6 study further evaluates the long-term safety and effectiveness of osilodrostat in patients with endogenous Cushing's syndrome during 3 years of routine clinical practice; data from the prespecified 2-year interim analysis are reported here

CONCLUSIONS

- **At the prespecified 2-year interim analysis cut-off, the safety profile of osilodrostat in patients with endogenous Cushing's syndrome was as expected and consistent with previous studies[2–5]**
 - **AEs related to osilodrostat were slightly more frequent in prior than in new osilodrostat users; no new safety signals were identified**
 - **The most common AE in new and prior osilodrostat users was adrenal insufficiency**
 - **Most AEs were mild or moderate in severity and manageable with corrective therapy**
 - **AEs related to hypocortisolism occured at similar frequencies in new and prior osilodrostat users; few led to permanent osilodrostat discontinuation**
 - **AEs related to accumulation of adrenal hormone precursors were more frequent in new versus prior users; however, none of these AEs led to osilodrostat discontinuation among new users**
 - **SAEs were more common in new than in prior osilodrostat users**
- **Rates of mUFC and LNSC normalization increased in new users from baseline to month 6 and were maintained in most prior users**
- **These LINC 6 interim data show the real-world clinical utility of osilodrostat for the management of endogenous Cushing's syndrome, providing cortisol normalization in both new and prior users with differing etiologies**

METHODS

- The ongoing prospective, non-interventional, multinational LINC 6 (NCT05382156) study enrolled the first adult patient with endogenous Cushing's syndrome on June 13, 2022, and will follow individual patients for up to 3 years
- Patients were enrolled across 40 sites in the USA and Europe (France, Germany, Italy, the Netherlands) where osilodrostat is approved and available
 - Patients could have received prior osilodrostat treatment either as monotherapy or in combination with another treatment for hypercortisolism
 - Patients were usually treated according to local prescribing information at the treating physician's discretion
 - Patients could not receive another investigational drug in another clinical trial
 - Patients who were treated with osilodrostat and consented to data collection were enrolled consecutively
- The primary endpoint is incidence of AEs and SAEs, with a focus on AEs related to hypocortisolism, accumulation of adrenal hormone precursors, arrhythmogenic potential and QT prolongation, and pituitary tumor enlargement
- Key secondary endpoints include normalization of mUFC, LNSC and serum cortisol levels among patients with assessments at baseline and month 6
- AEs were recorded at each patient visit
- Outcomes were assessed separately in new (those starting osilodrostat at or after study entry) and prior osilodrostat users (those starting osilodrostat at any time before study entry) at the prespecified 2-year interim cut-off (July 24, 2024); all assessments are descriptive

RESULTS

Baseline characteristics

- Overall, 206 patients were included in the safety population (received ≥1 osilodrostat dose); osilodrostat treatment status at enrollment was available for 205 patients
- Of the 205 patients evaluated, **70** were **new** and **135** were **prior** osilodrostat users
- Patient characteristics were typical of a population with Cushing's syndrome and were mostly similar between new and prior osilodrostat users; however, a greater proportion of prior users had Cushing's disease
- Baseline mUFC was just above the ULN in prior users but substantially higher in new osilodrostat users

Country N (%)	France 84 (41.0)	USA 81 (39.5)	Germany 27 (13.2)	Italy 8 (3.9)	Netherlands 5 (2.4)
New osilodrostat users: n (%)	32 (45.7)	16 (22.9)	12 (17.1)	5 (7.1)	5 (7.1)
Prior osilodrostat users: n (%)	52 (38.5)	65 (48.1)	15 (11.1)	3 (2.2)	0

Patient characteristics	New osilodrostat users (n=70)	Prior osilodrostat users (n=135)
Mean (SD) age, years	50.8 (15.1)	54.1 (13.9)
Female, n (%)	55 (78.6)	100 (74.1)
Mean (SD) weight, kg	84.1 (18.6)	84.7 (26.8)
Mean (SD) BMI, kg/m²	30.8 (6.9)	30.8 (9.9)
Cushing's disease, n (%)	49 (70.0)	112 (83.0)
Non-pituitary Cushing's syndrome, n (%)	**21 (30.0)**	**23 (17.0)**
Ectopic	4 (5.7)	13 (9.6)
Adrenal adenoma	9 (12.9)	3 (2.2)
BND	5 (7.1)	5 (3.7)
Adrenal carcinoma	2 (2.9)	0
Other*	1 (1.4)	2 (1.5)
Patients with prior surgery, n (%)	25 (59.5)†	86 (63.7)
Mean (SD) mUFC, nmol/24 h	587.5 (723.6)	148.9 (152.1)

*Other refers to primary pigmented nodular adrenocortical disease (**n=1**) in **new** users and micronodular adrenal dysplasia and unknown (both **n=1**) in **prior** users; †Data were available for only 42 patients

Osilodrostat dose and exposure

Median (min–max) and mean (SD) on study:

	New users	Prior users
Dose	**4.7** mg/day (1.6–69.3); **9.6** mg/day (13.0)	**5.0** mg/day (0.5–80.0); **9.9** mg/day (12.2)
Exposure	**4.2** months (0.4–19.6); **5.2** months (4.3)	**10.4** months (0–20.8); **10.8** months (5.3)

1. The proportions of patients experiencing an AE were similar among new and prior users; AEs related to osilodrostat were slightly more frequent in prior users

	New users	Prior users
≥1 AE:	51.4% (n=36/70)	54.1% (n=73/135)
≥1 AE related to osilodrostat:	17.1% (n=12/70)	23.0% (n=31/135)
Number of AEs related to osilodrostat:	30	75

Most common AEs (≥6.0% of events):

- New users: Adrenal insufficiency 16.7% (n=5/30); Gastrointestinal disorder 6.7% (n=2/30); Hypokalemia 6.7% (n=2/30); Headache 6.7% (n=2/30); Abnormal hair growth 6.7% (n=2/30)
- Prior users: Adrenal insufficiency 13.3% (n=10/75); Dizziness 8.0% (n=6/75); Diarrhea 6.7% (n=5/75); Fatigue 6.7% (n=5/75)

- Most treatment-related AEs were mild or moderate in both **new** (**93.3%**) and **prior** (**94.7%**) osilodrostat users
- AEs led to death in two **new** users (**2.9%**, n=2/70; adrenal insufficiency, adrenocortical carcinoma and lung disorder) and three **prior** osilodrostat users (**2.2%**, n=3/135; hemolysis, hypovolemic shock after probable adrenal insufficiency, sepsis and respiratory distress)

2. Most AEs were managed by corrective therapy,* dose adjustments or interruption

- Six **new** (**8.6%**, n=6/70) and 11 **prior** (**8.1%**, n=11/135) users discontinued because of 21 AEs (**new**, n=6; **prior**, n=15)

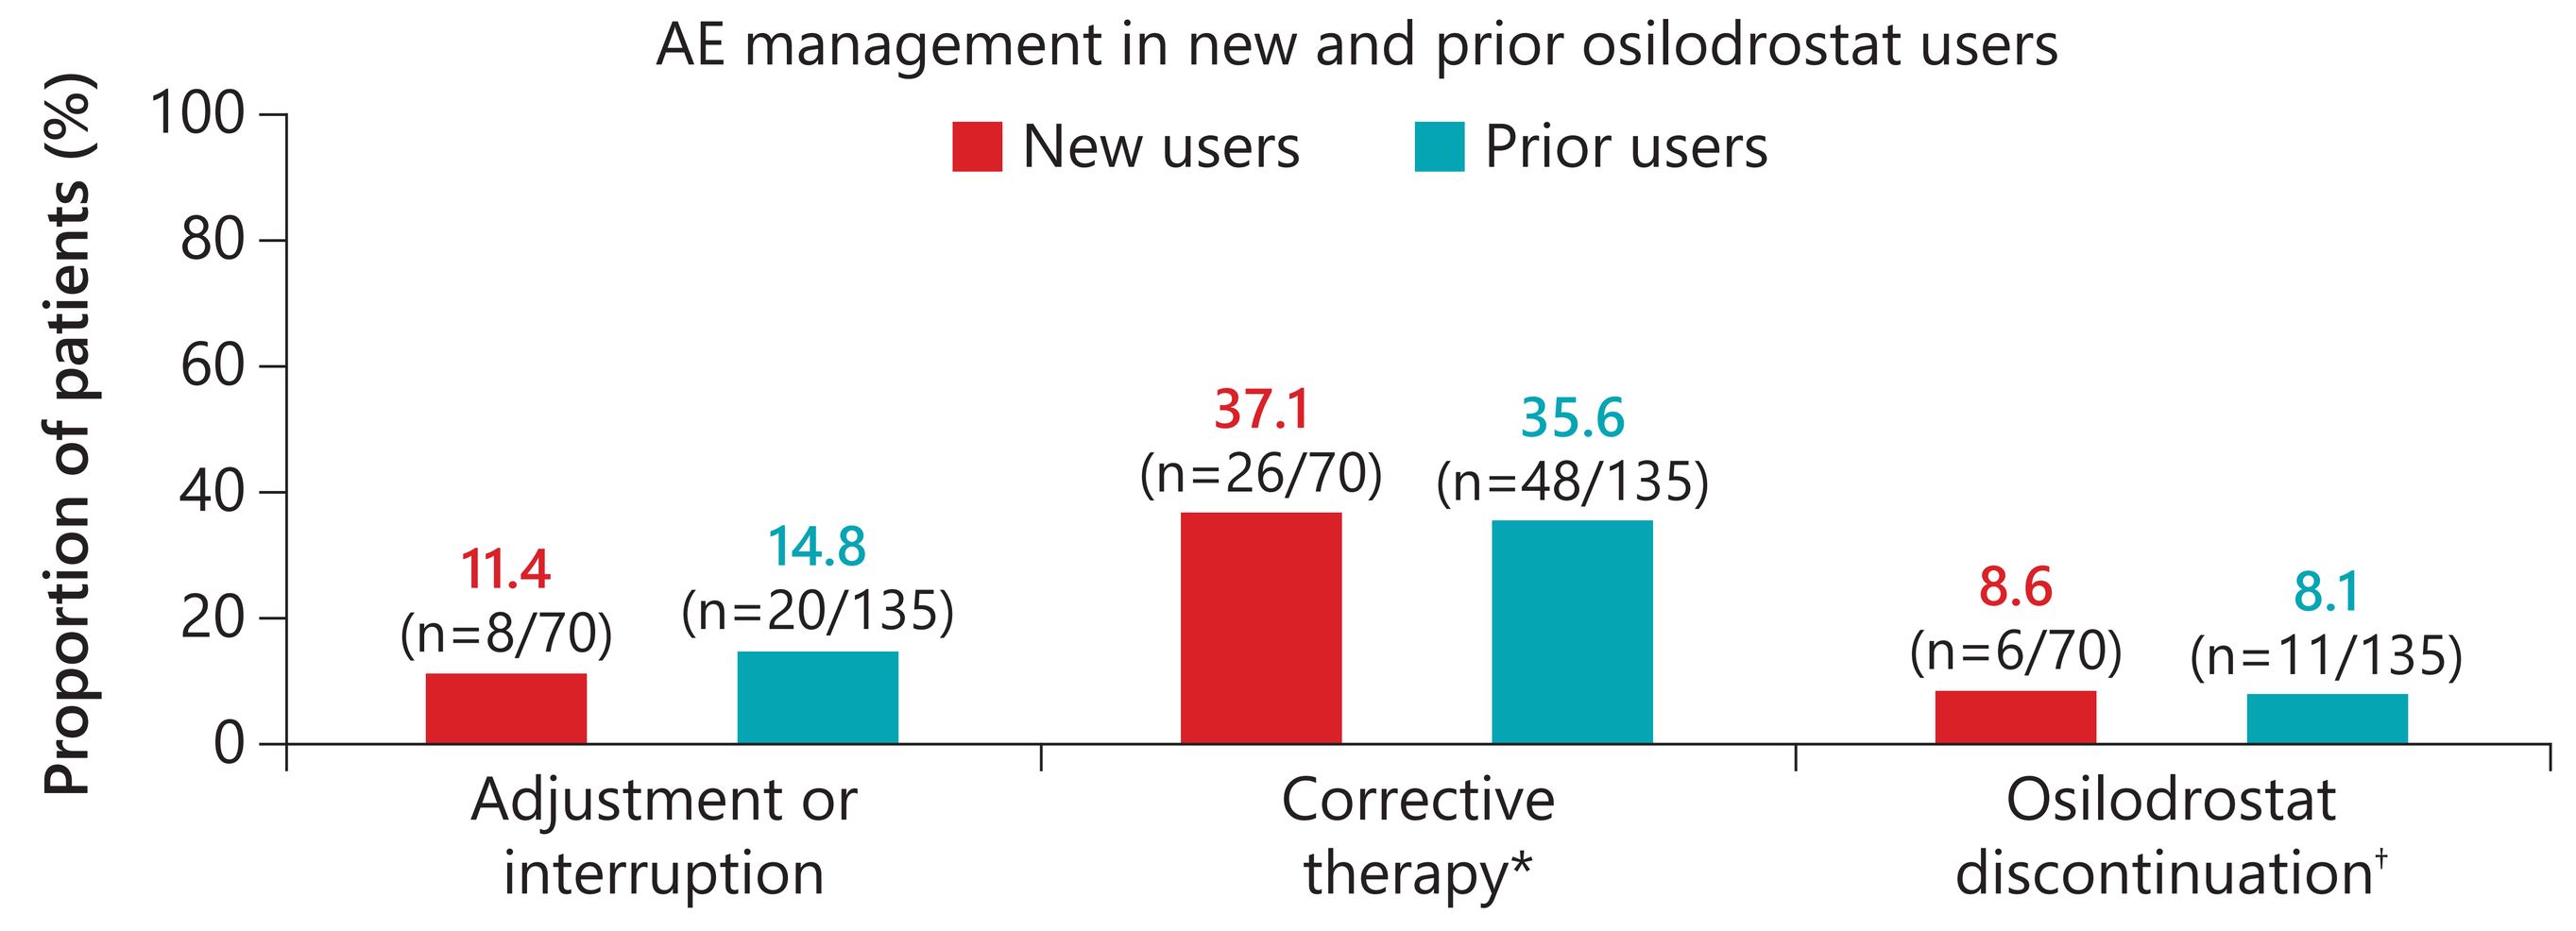
AE management in new and prior osilodrostat users
New users
Prior users
Proportion of patients (%)
100
80
60
40
20
0
11.4
(n=8/70)
14.8
(n=20/135)
37.1
(n=26/70)
35.6
(n=48/135)
8.6
(n=6/70)
8.1
(n=11/135)
Adjustment or interruption
Corrective therapy*
Osilodrostat discontinuation†

*Includes concomitant medication (including glucocorticoids), surgery and other (blood transfusion, blood letting, scans, antibiotics or catecholamines, percutaneous drain placement, intravenous fluids, total parenteral nutrition, parenteral hydration, nasogastric tube, ventilation, urinary catheter, emergency room visit outside of hospital); **New** users also reported adrenal insufficiency, hyperadrenocorticism, adrenal gland cancer, adrenocortical carcinoma, pregnancy and acute pulmonary edema (all n=1/6; **16.7%** each); **prior** users reported adrenal insufficiency (n=2/11; **18.2%**), hyperandrogenism, nausea, vomiting, disease progression, sepsis, abnormal hormone level, benign pituitary tumor, dizziness, pregnancy, adrenalectomy, inadequately controlled blood pressure, hypertension and orthostatic hypotension (all n=1/11; **9.1%** each)

- One **new** (**1.4%**, n=1/70) and six **prior** (**4.4%**, n=6/135) users discontinued because of 11 AEs (**new**, n=1; **prior**, n=10) considered treatment related

3. AEs related to hypocortisolism or accumulation of adrenal hormone precursors were infrequent and manageable without discontinuation in most patients

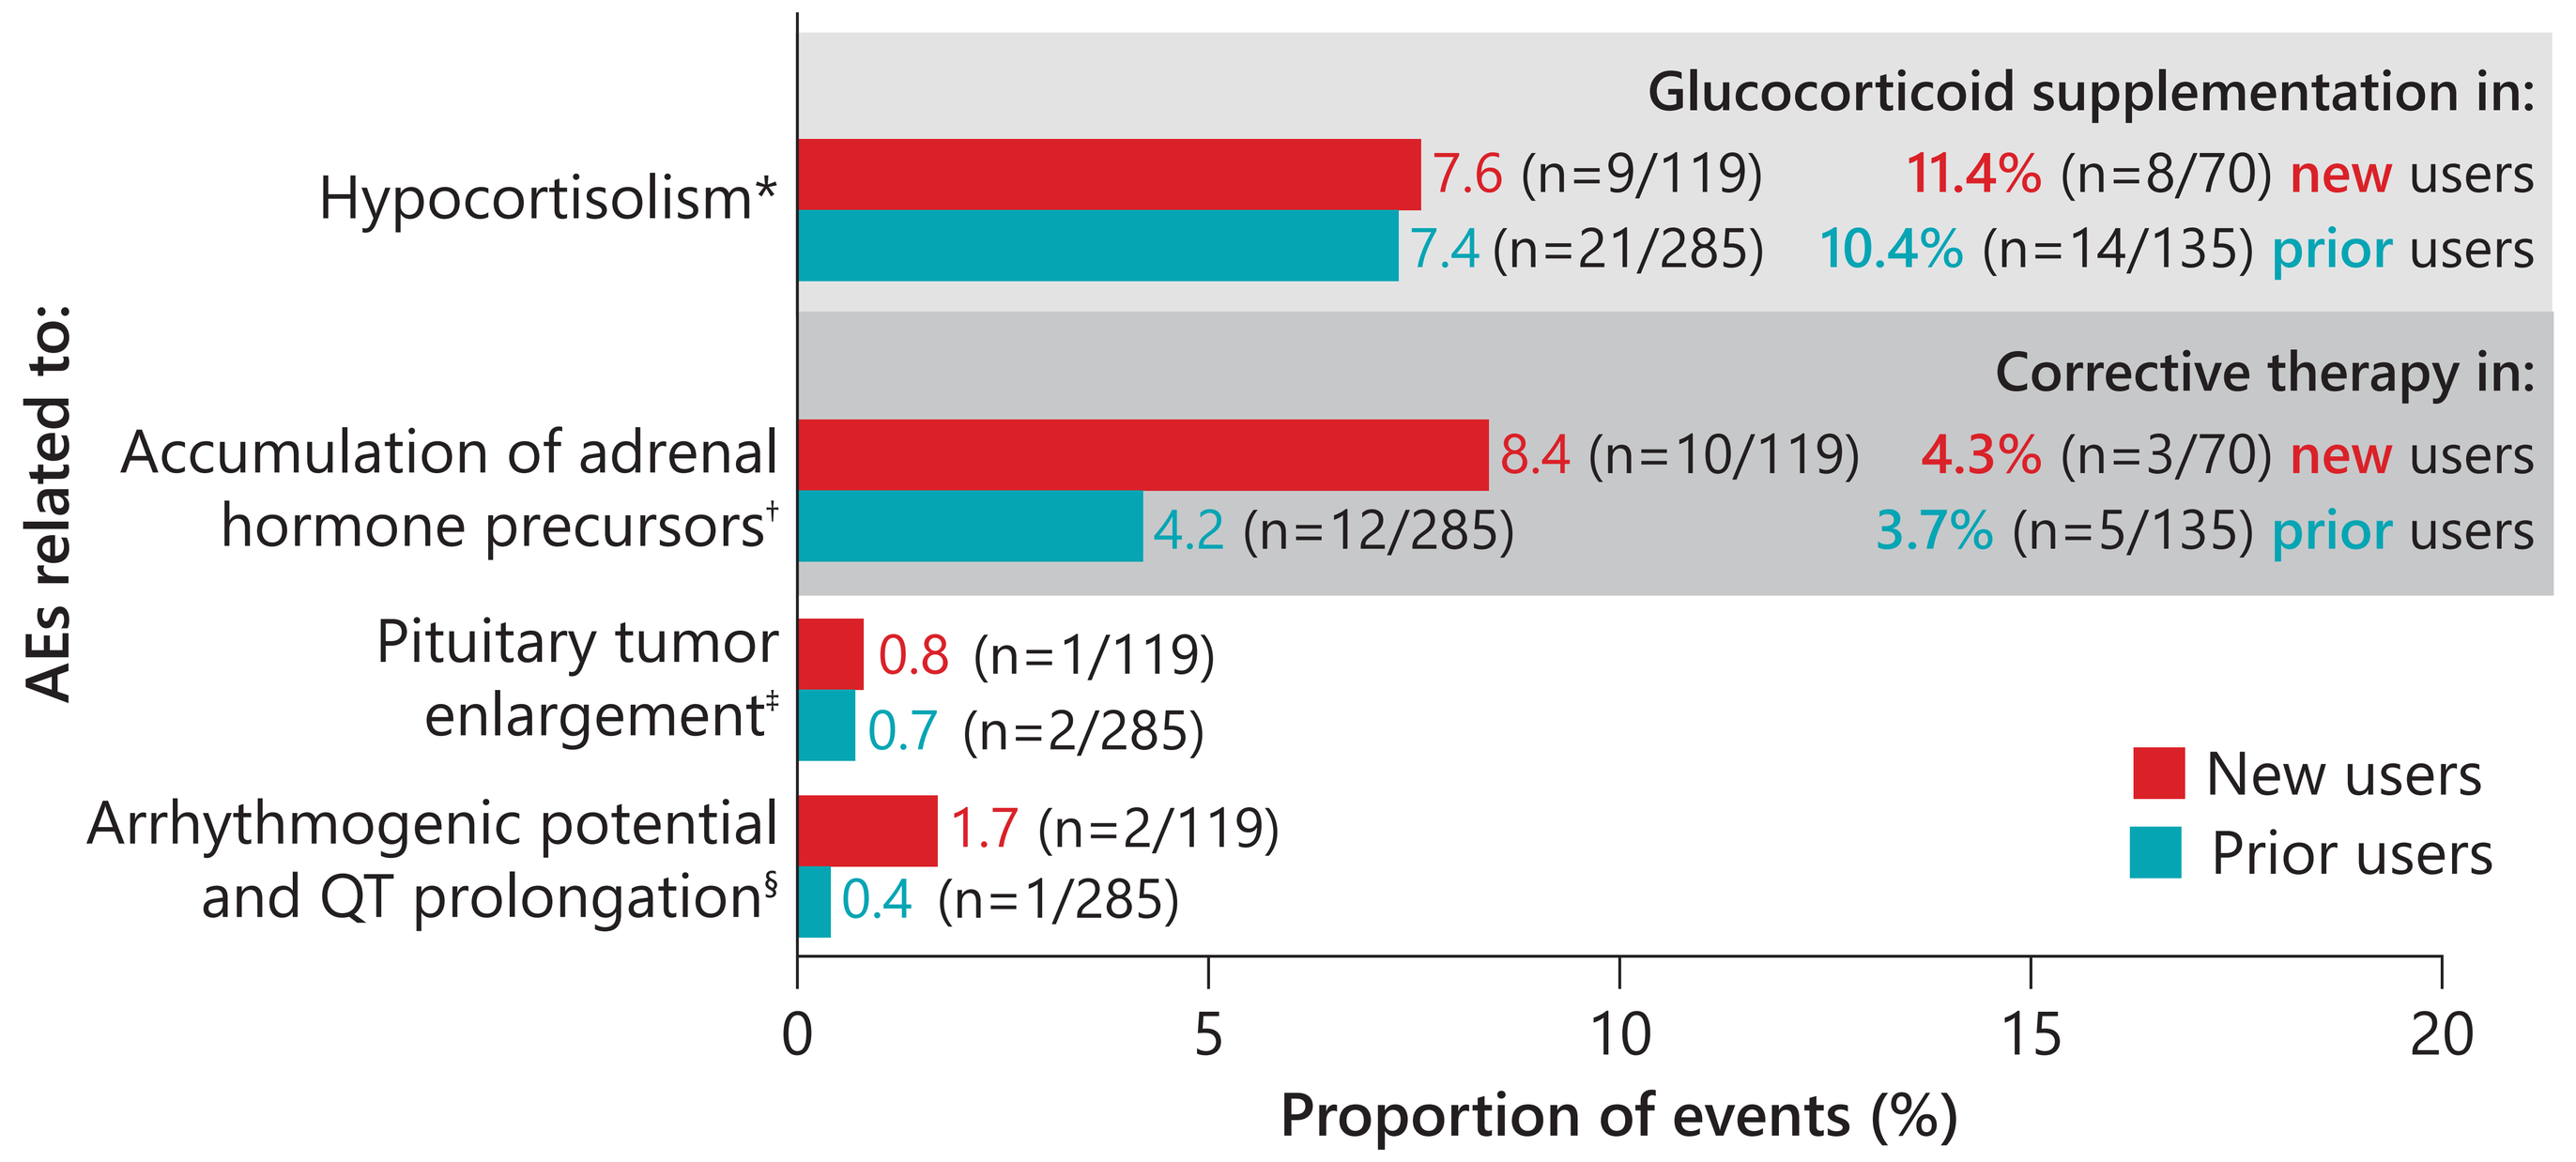
AEs related to:
Glucocorticoid supplementation in:
Hypocortisolism*
7.6 (n=9/119)
7.4 (n=21/285)
11.4% (n=8/70) new users
10.4% (n=14/135) prior users
Corrective therapy in:
Accumulation of adrenal hormone precursors†
8.4 (n=10/119)
4.2 (n=12/285)
4.3% (n=3/70) new users
3.7% (n=5/135) prior users
Pituitary tumor enlargement‡
0.8 (n=1/119)
0.7 (n=2/285)
Arrhythmogenic potential and QT prolongation§
1.7 (n=2/119)
0.4 (n=1/285)
New users
Prior users
0
5
10
15
20
Proportion of events (%)

Reported as (events): *Adrenal insufficiency (new=6; prior=16), acute adrenocortical insufficiency (new=2; prior=4) and glucocorticoid deficiency (new=1; prior=1); †Peripheral edema (new=2; prior=3), hypokalemia (new=2; prior=2), abnormal hair growth (new=3; prior=0), acne (new=2; prior=1), hirsutism (new=1; prior=2), hyperandrogenism (new=0; prior=2) and hypertension (new=0; prior=2); ‡ACTH-producing pituitary tumor (new=1; prior=0), pituitary tumor (new=0; prior=1) and benign pituitary tumor (new=0; prior=1); §Loss of consciousness (new=1; prior=0) and syncope (new=1; prior=1)

- Few AEs related to hypocortisolism (**new** user, **11.1%** of events [n=1/9]; **prior** user, **9.5%** of events [n=2/21]) or accumulation of adrenal hormone precursors (**new** user, **0%** of events; **prior** user, **16.7%** of events [n=2/12]) led to discontinuation of osilodrostat
- Similar proportions of patients in each group required glucocorticoid supplementation for hypocortisolism or corrective therapy for accumulation of adrenal hormone precursors

4. SAEs occurred at a higher frequency in new than in prior osilodrostat users

20.0% (n=14/70) of new users experienced an SAE
16.7% (n=4/24) were treatment related

9.6% (n=13/135) of prior users experienced an SAE
33.3% (n=7/21) were treatment related

Most commonly (>1 occurrence):

- Acute kidney injury (12.5%, n=3/24)
- Hypokalemia (8.3%, n=2/24)
- Adrenal insufficiency (8.3%, n=2/24)
- Pneumonia (8.3%, n=2/24)
- Adrenocortical insufficiency (19.0%, n=4/21)

New users also reported acute adrenocortical insufficiency, hyperadrenocorticism, intestinal diverticulitis perforation, asthenia, pyrexia, hematoma infection, acute pyelonephritis, dehydration, hyperkalemia, ACTH-producing pituitary tumor, adrenocortical carcinoma, breast cancer, loss of consciousness, acute pulmonary edema and lung disorder (all n=1/24, **4.2%** each); **prior** users reported hemolysis, adrenal insufficiency, death, disease progression, infection, bacterial pneumonia, viral pneumonia, sepsis, septic shock, hyperkalemia, glioblastoma, pituitary tumor, benign pituitary tumor, altered state of consciousness, acute kidney injury, asthma and respiratory distress (all n=1/21, **4.8%** each)

5. Rates of mUFC and LNSC normalization increased from baseline to month 6 in new users and were maintained in most prior users

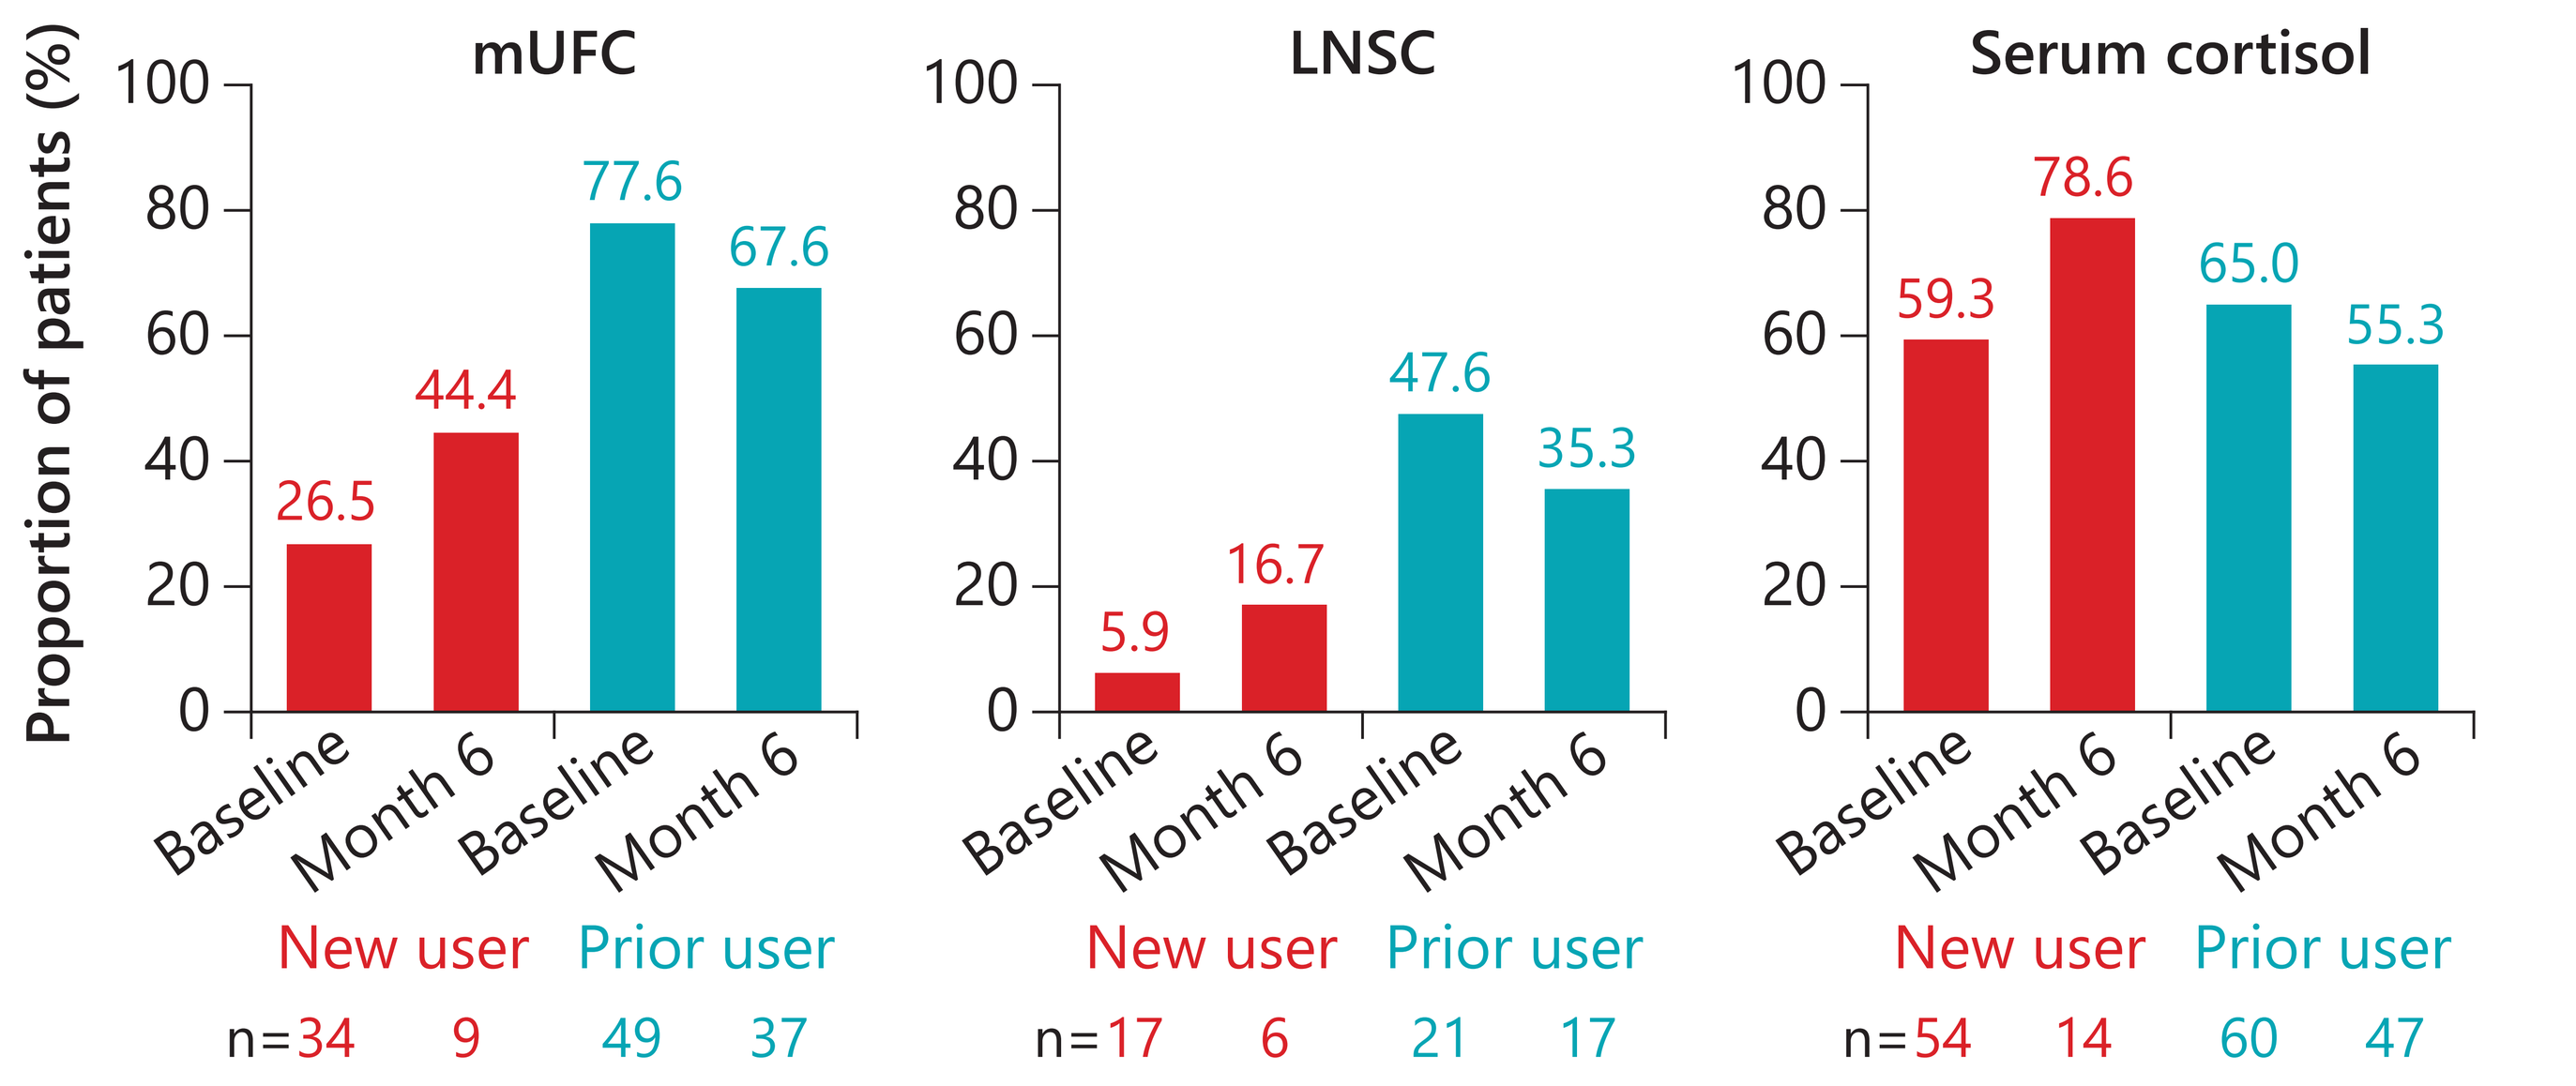
Proportion of patients (%)
mUFC
26.5
44.4
77.6
67.6
LNSC
5.9
16.7
47.6
35.3
Serum cortisol
59.3
78.6
65.0
55.3
Baseline
Month 6
Baseline
Month 6
New user
Prior user
n=34 9 49 37
n=17 6 21 17
n=54 14 60 47

Normal values were defined by each center. Serum cortisol levels were assessed according to standard clinical practice at each site, but this was not mandated by the study protocol. At the prespecified 2-year interim analysis cut-off, not all patients had had their 6-month visit; therefore, patient numbers were small for some analyses

ACKNOWLEDGMENTS

We thank Lyssa Gold of AMICULUM Business Services Ltd (funded by Recordati AG) for providing medical editorial assistance, as well as the site investigators, study coordinators/nurses and patients who are participating in the study.

DISCLOSURES

The LINC 6 study is sponsored by Recordati AG.

ABBREVIATIONS

ACTH, adrenocorticotropic hormone; AE, adverse event; BMI, body mass index; BND, bilateral adrenal nodular disease; LNSC, late-night salivary cortisol; mUFC, mean urinary free cortisol; SAE, serious adverse event; SD, standard deviation; ULN, upper limit of normal

REFERENCES

1. Fleseriu M *et al. Pituitary* 2022;25:959–70; 2. Pivonello R *et al. Lancet Diabetes Endocrinol* 2020;8:748–61;
3. Fleseriu M *et al. Eur J Endocrinol* 2022;187:531–41; 4. Gadelha M *et al. Front Endocrinol (Lausanne)* 2023;14:1236465;
5. Gadelha M *et al. J Clin Endocrinol Metab* 2022;107:e2882–95; 6. Tanaka T *et al. Endocr J* 2020;67:841–52

PRESENTED AT: The American Association of Clinical Endocrinology Annual Meeting, Orlando, FL, USA, May 15–17, 2025