HEMOLYTIC MARKERS, MORTALITY, AND THROMBOEMBOLIC EVENTS IN COLD AGGLUTININ DISEASE (CAD): A RETROSPECTIVE ANALYSIS OF THE OPTUM® DE-IDENTIFIED ELECTRONIC HEALTH RECORD DATASET IN THE UNITED STATES

Catherine Broome[1]; Wilma Barcellini[2]; Alexander Röth[3]; Alia Karaouni[4]; Florence Joly[5]; Ronnie Yoo[6]; Augustin Terlinden[5]; Ayo Adeyemi[7]; Yola Moride[8]; Quentin A. Hill[9]

[1]Division of Hematology, MedStar Georgetown University Hospital, Washington, DC, United States; [2]Hematology Unit, Fondazione IRCCS Ca' Granda Ospedale Maggiore Policlinico, Milan, Italy; [3]Department of Hematology and Stem Cell Transplantation, West German Cancer Center, University Hospital Essen, Essen, Germany; [4]BMAPS SARL, Geneva, Switzerland; [5]Sanofi, Chilly-Mazarin, France; [6]Sanofi, Boston, MA, United States; [7]Sanofi, Cambridge, MA, United States; [8]Yolarx Consultants, Inc., Montreal, Quebec, Canada; [9]Department of Clinical Haematology, Leeds Teaching Hospitals NHS Trust, Leeds, United Kingdom

INTRODUCTION

- Cold agglutinin disease (CAD) is a rare autoimmune hemolytic anemia (AIHA) characterized by the destruction of red blood cells, which is mediated by activation of the classical complement pathway.[1]
- Although there is consensus on an increased risk of thromboembolic events (TEs) in patients with CAD,[2] the evidence for the effect on mortality is mixed.
- Recent studies support an increased risk of mortality in patients with CAD in Denmark and the US.[3–5]

AIMS

- The aims of this retrospective study were:
 1. To evaluate whether patients with CAD in the US have a higher risk of mortality and TEs compared with a matched non-CAD population.
 2. To determine the association between CAD biomarkers and the risk of mortality and TEs in patients with CAD.

METHODS

Study design and participants

- This was a retrospective, cohort study of patients with CAD and exact matched patients without CAD in the US, identified using the Optum® de-identified Electronic Health Record dataset (2007–2021).
- Patients in the non-CAD cohort had no evidence of AIHA-related diagnosis codes and no mention of CAD in their records.

Assessment of mortality, thromboembolic events, and biomarkers

- Mortality and TEs were assessed over the follow-up period (from index date to end of medical activity, study period, or death).
- Biomarker levels were assessed at baseline and during follow-up, and categorized as follows:
 - Bilirubin: elevated, >1.2 mg/dL; normal, ≤1.2 mg/dL
 - Lactate dehydrogenase (LDH): elevated, >250 U/L; normal, ≤250 U/L
 - Hemoglobin (Hb): no anemia, Hb ≥12 g/dL; mild, Hb ≥10 g/dL and <12 g/dL; moderate, Hb ≥8 g/dL and <10 g/dL; and severe, Hb <8 g/dL.

Statistical analyses

- Adjusted ratios presented in the following aims account for age, sex, race, geographic region, as well as confounding variables, such as smoking, comorbidities (Charlson comorbidity index), past TEs, and index season.

Aim 1: Risk of mortality or TEs in patients with CAD compared with a non-CAD cohort

- Adjusted incidence rate ratios (aIRRs) and 95% confidence intervals (CIs) for mortality and TEs were obtained using multivariate Poisson regressions.
- Probabilities of survival and remaining TE-free were assessed using Kaplan–Meier analysis with log-rank testing.
- Adjusted hazard ratios (aHRs) and 95% CIs were obtained using multivariate Cox proportional hazards regression analysis.

Aim 2: Association between biomarkers and the risk of mortality and TEs in patients with CAD

- Analyses were conducted within the CAD cohort to assess the correlation between severity of hemolysis and anemia as measured by CAD biomarkers (all observations until event) and risk of mortality or TE.
- Adjusted hazard ratios (aHRs) and 95% CIs were obtained using time-varying Cox regressions.

REFERENCES

1. Berentsen S. *Hematology Am Soc Hematol Educ Program* 2016;2016(1):226–31.; 2. Broome CM, et al. *Res Pract Thromb Haemost* 2020;4(4):628–35.; 3. Hansen DL, et al. *Eur J Haematol* 2022;109:10–20.; 4. Bylsma L, et al. *Blood Adv* 2019;3:2980–5.; 5. Hill QA, et al. *Blood* 2019;134:S1:4790.

RESULTS

Table 1. Baseline demographics and clinical characteristics of patients with and without CAD in the US

Characteristic	CAD cohort (N=457)	Non-CAD cohort (N=2,285)	*p*-value
Mean age at index date (SD), years	66.82 (16.16)	66.82 (16.15)	1.000
Female, n (%)	286 (63)	1430 (63)	1.000
Race, n (%)			1.000
White	404 (88)	2020 (88)	
Black/African American	24 (5)	120 (5)	
Other	29 (6)	145 (6)	
Geographic region, n (%)			1.000
Northeast	58 (13)	290 (13)	
Midwest	251 (55)	1255 (55)	
South	90 (20)	450 (20)	
West	41 (9)	205 (9)	
Unknown	17 (4)	85 (4)	
Medical history, n (%)			
Any TE	67 (14.7)	104 (4.6)	<0.001
Received blood transfusion	67 (14.7)	<11 (<1.0)	<0.001
Weight baseline, kg			
Mean (SD)	76.33 (19.33)	79.05 (19.73)	0.0251
Data unavailable, n (%)	100 (21.9)	1293 (56.6)	
Smoking status, n (%)			<0.001
Current smoker	124 (27.1)	286 (12.5)	
Past smoker	162 (35.4)	416 (18.2)	
Never smoked	41 (9.0)	135 (5.9)	
Other, unknown, missing	130 (28.4)	1448 (63.4)	
CCI, n (%)			<0.001
0	174 (38.1)	1525 (66.7)	
1	89 (19.5)	281 (12.3)	
2	61 (13.3)	181 (7.9)	
≥3	133 (29.1)	298 (13.0)	

Exact matching was used to generate a sample of non-CAD patients who were matched to the CAD patients on key characteristics (age, sex, race, geographic region, and index year). Baseline was 12 months prior to index date. CAD, cold agglutinin disease; CCI, Charlson Comorbidity Index; SD, standard deviation; TE, thromboembolic event.

Table 2. Risk of mortality or TEs in patients with CAD compared with a non-CAD cohort

CAD / non-CAD	Mortality		TE	
	Ratio (95% CI)	*p*-value	Ratio (95% CI)	*p*-value
aIRR	2.19 (1.78; 2.67)	0.001	2.24 (1.83; 2.72)	0.001
aHR	2.21 (1.81; 2.71)	0.001	2.13 (1.75; 2.59)	0.001

Analyses were adjusted for age, sex, race, geographic region as well as confounding variables, such as smoking, comorbidities, past TEs, and index season. aIRR, adjusted incidence rate ratio; aHR, adjusted hazard ratio; CI, confidence interval; TE, thromboembolic event

- The adjusted incidence rate ratio and hazard ratio of death in the CAD cohort was twice that of the non-CAD cohort.
- Similarly, adjusted TE–related risk ratios were doubled among the CAD patients compared with the non-CAD group.

Figure 1. Survival probability for patients with CAD compared with a matched cohort of patients without CAD

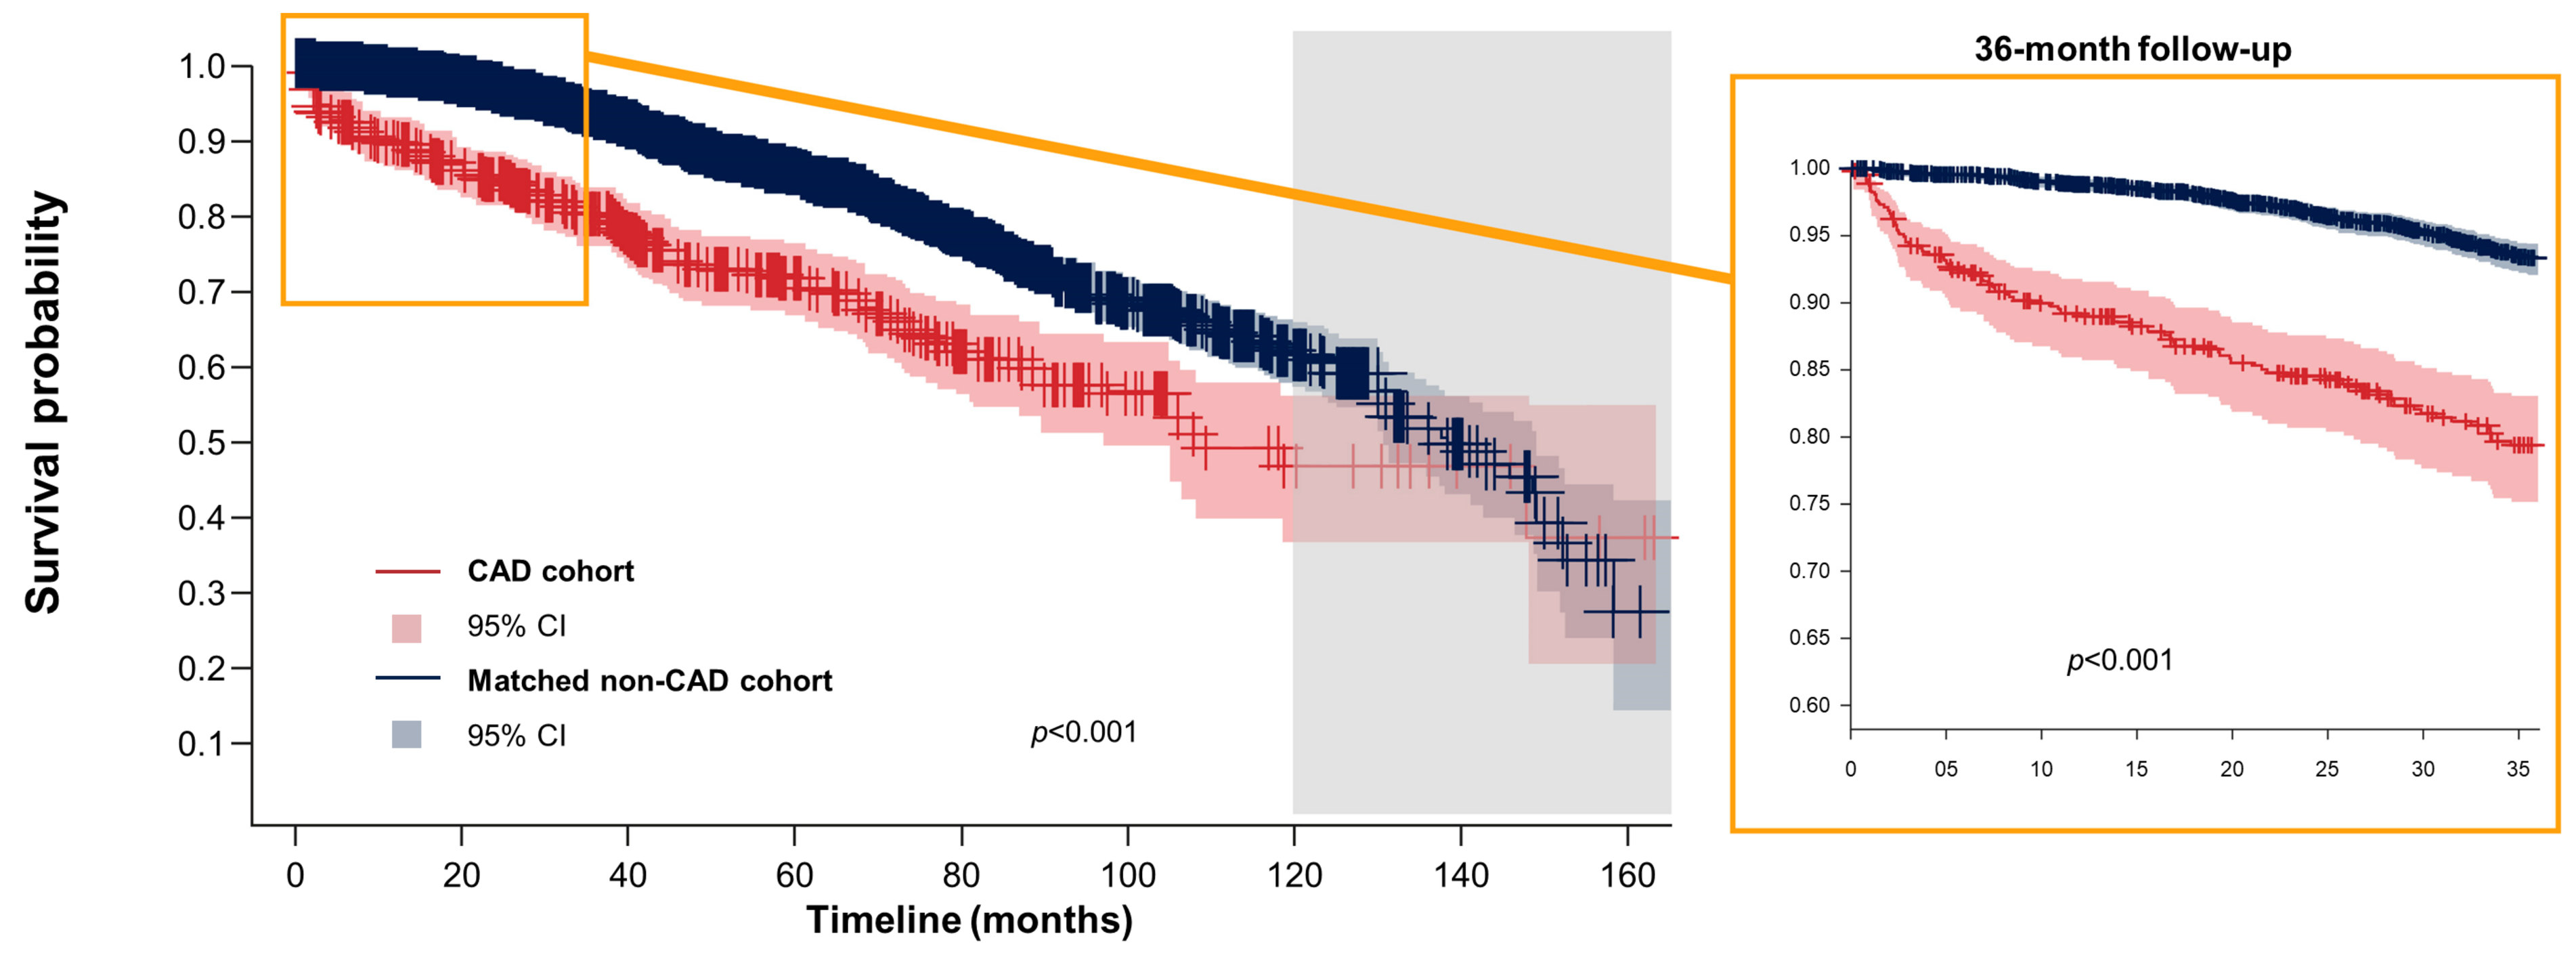
Survival probability
Timeline (months)
CAD cohort
95% CI
Matched non-CAD cohort
95% CI
p<0.001
36-month follow-up
p<0.001

	0	20	40	60	80	100	120	140	160
CAD cohort									
At risk	457	339	233	155	85	41	17	6	3
Events	0	63	92	108	125	131	136	136	137
Matched non-CAD cohort									
At risk	2285	1900	1374	893	525	254	117	45	3
Events	0	49	146	226	296	343	362	377	386

p-values were assessed using the log-rank test. Survival probability after 120 months (greyed out) is unreliable as a result of low patient numbers.
CAD, cold agglutinin disease; CI, confidence interval

Figure 2. Probability to remain TE-free for patients with CAD compared with a matched cohort of patients without CAD

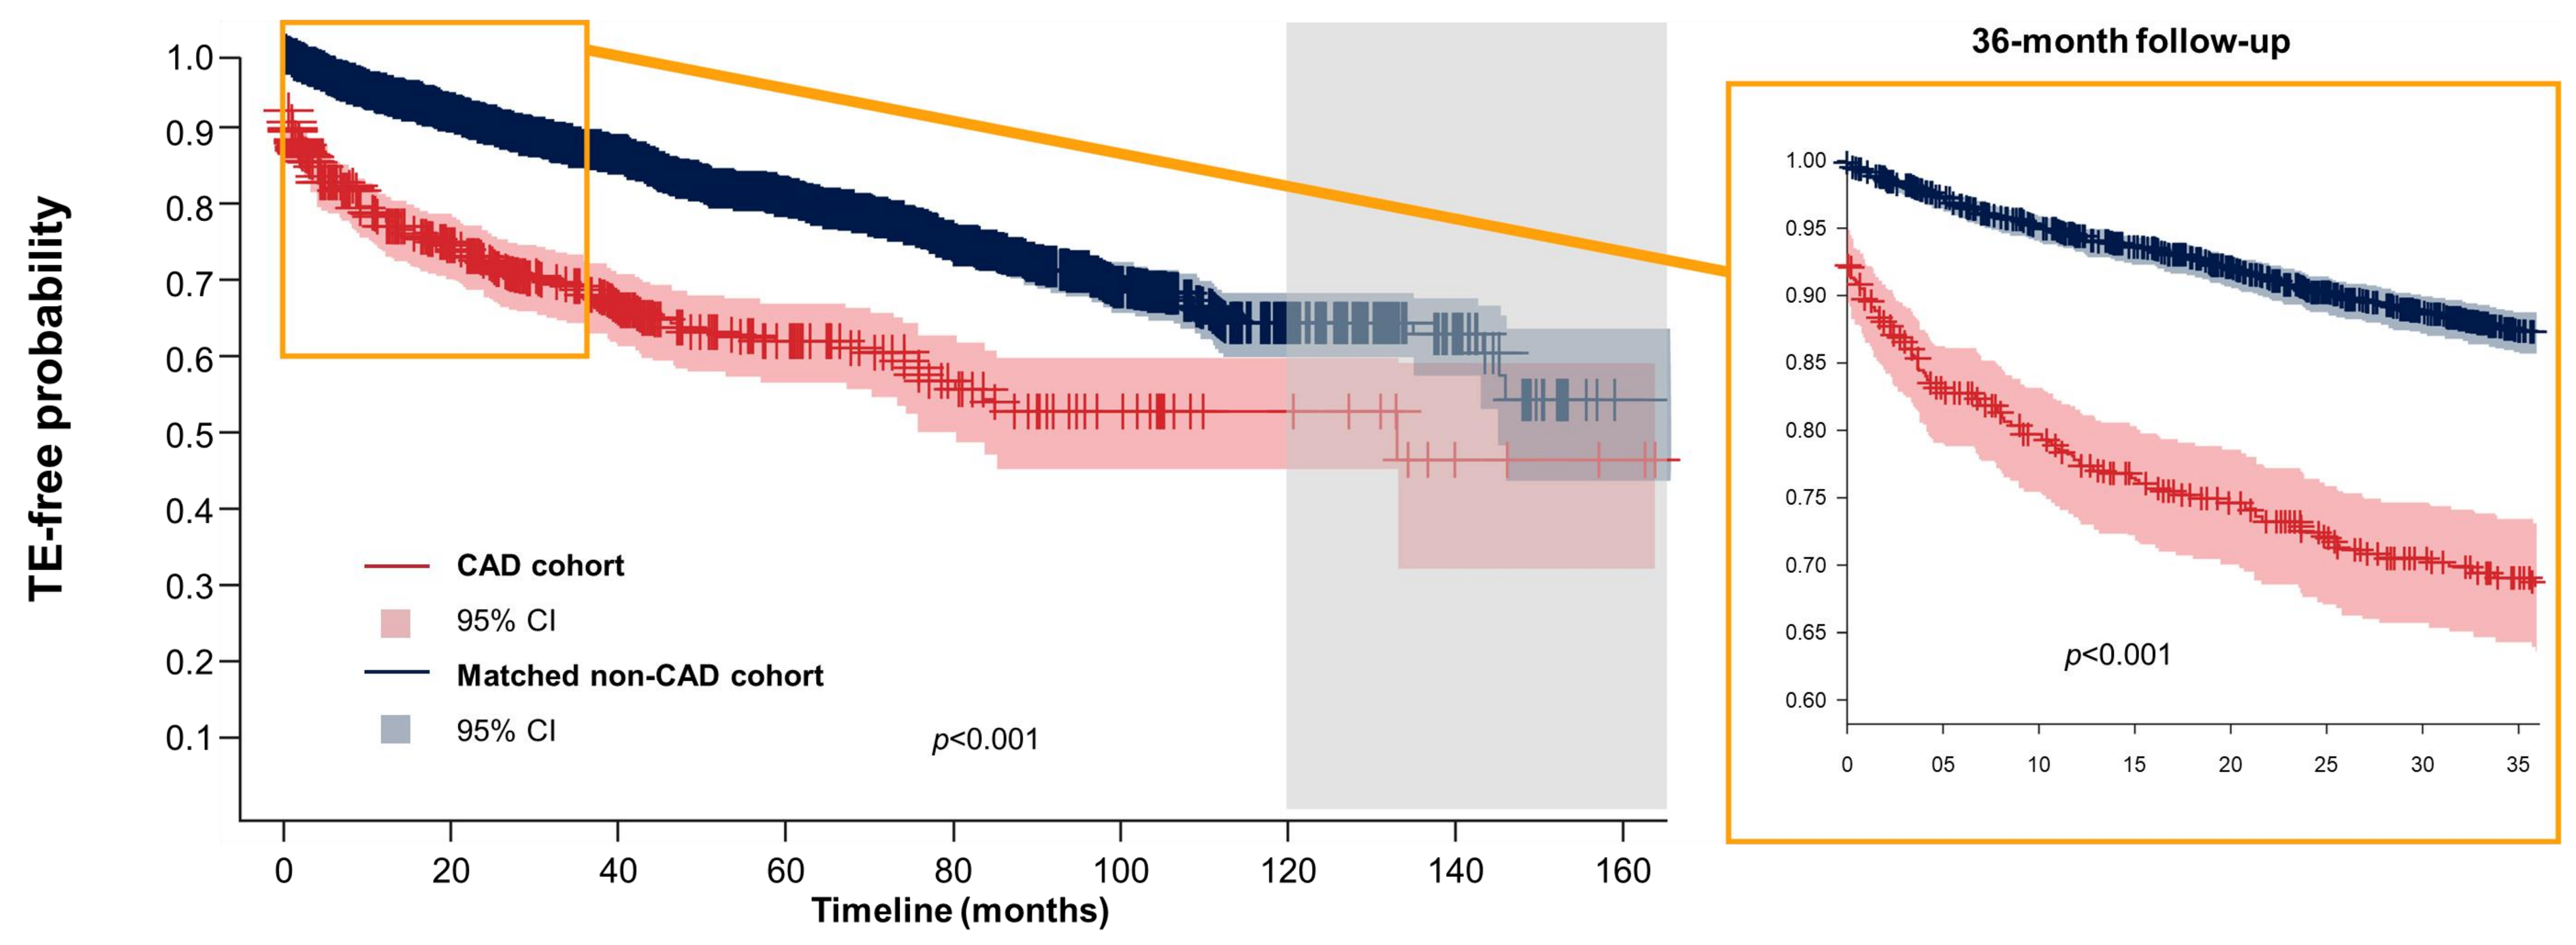
TE-free probability
Timeline (months)
CAD cohort
95% CI
Matched non-CAD cohort
95% CI
p<0.001
36-month follow-up
p<0.001

	0	20	40	60	80	100	120	140	160
CAD cohort									
At risk	433	265	162	99	50	23	12	4	2
Events	24	109	134	143	149	152	152	153	153
Matched non-CAD cohort									
At risk	2283	1756	1205	747	420	194	83	36	1
Events	2	172	266	330	375	397	407	408	411

p-values were assessed using the log-rank test. TE-free probability after 120 months (greyed out) is unreliable due to low patient numbers.
CAD, cold agglutinin disease; CI, confidence interval; TE, thromboembolic event

- Overall, survival probability was significantly lower in the CAD cohort than in the non-CAD cohort during full follow-up (**Figure 1**), and at 12-, 24-, and 36-months (all *p*<0.001).
- Similarly, probability to remain TE-free was significantly lower for patients with CAD than those without during full follow-up (**Figure 2**), and at 12-, 24-, and 36-months (all p<0.001).

Table 3. Mortality and TEs by biomarker state in the CAD cohort

Biomarker state (compared with normal)	Mortality		TE	
	aHR (95% CI)	*p*-value	aHR (95% CI)	*p*-value
Hemoglobin: mild anemia	2.71 (1.48; 4.95)	0.001	1.37 (0.82; 2.29)	0.235
Hemoglobin: moderate anemia	7.23 (4.12; 12.69)	0.001	2.19 (1.28; 3.72)	0.001
Hemoglobin: severe anemia	17.78 (9.72; 32.51)	0.001	3.84 (2.01; 7.35)	0.001
Elevated bilirubin	1.59 (1.10; 2.30)	0.013	1.32 (0.86; 2.04)	0.206
Elevated LDH	1.44 (0.96; 2.17)	0.076	1.19 (0.73; 1.94)	0.490

All observations until the event were included in the analyses. Analyses were adjusted for age, sex, race, geographic region as well as confounding variables, such as smoking, comorbidities, past TEs, and index season. Hazard ratios compare the biomarker state to the normal state for each biomarker: for anemia, no anemia; for bilirubin, normal bilirubin; for LDH, normal LDH.
aHR, adjusted hazard ratio; CI, confidence interval; LDH, lactate dehydrogenase; TE, thromboembolic event

- In the CAD cohort, patients with mild, moderate, or severe anemia had a significantly higher risk of mortality than those without anemia (all *p*<0.001).
- Moderate and severe anemia were also associated with increased risk of TEs compared with no anemia (both *p*<0.001).
- Elevated bilirubin levels were significantly associated with an increased risk of mortality compared with normal levels (*p*=0.013).
- Elevated LDH was somewhat associated with an increased risk of mortality but did not reach statistical significance (*p*=0.076).

CONCLUSIONS

- The clinical burden associated with CAD is significant and extends beyond anemia; patients with CAD were two times more likely to die or experience TEs than those without CAD
- Within the CAD cohort, the degree of ongoing hemolysis, as measured by increased bilirubin, and subsequent anemia were associated with an increased mortality risk; moderate and severe anemia were also associated with an increased risk of a TE.
- Chronic control of complement activation and the resulting hemolysis in CAD may therefore help manage the risk of mortality and TEs.

ACKNOWLEDGEMENTS AND FUNDING

Medical writing support for the development of this abstract, under the direction of the authors, was provided by Amy Watkins, PhD, of Ashfield MedComms, an Inizio company, and funded by Sanofi in accordance with Good Publication Practice guidelines.

This study was sponsored by Sanofi.